Immunological control by PARP inhibitors for successful immunotherapy of ovarian carcinoma

Holicek P[1,2], Vosahlikova S[1], Moserova I[1], Orsulic S[3], Mikyskova R[4], Hensler M[1], Kasikova L[1], Lanickova T[1], Novotny O[4], Reinis M[4], Drozenova J[5], Ryska A[6], Laco J[6], Rob L[7], Michal J. Halaska[7], Cibula D[8], Spisek R[1,2], Fucikova J[1,2]

[1]SOTIO Biotech, Prague, Czech Republic; [2]Department of Immunology, Charles University, 2nd Faculty of Medicine and University Hospital Motol, Prague, Czech Republic; [3]Department of Obstetrics and Gynecology, David Geffen School of Medicine, University of California, Los Angeles, CA 90095, USA; VA Greater Los Angeles Healthcare System, Los Angeles, CA 90073, USA; [4]Laboratory of Immunological and Tumour Models, Institute of Molecular Genetics of the Czech Academy of Sciences, Prague, Czech Republic; [5]Department of Pathology 3rd Faculty of Medicine and University Hospital Kralovske Vinohrady, Prague, Czech Republic; [6]The Fingerland Department of Pathology, Charles University, Faculty of Medicine and University Hospital Hradec Kralove, Czech Republic; [7]Department of Gynecology and Obstetrics, Charles University, 3rd Faculty of Medicine and University Hospital Kralovske Vinohrady, Prague, Czech Republic; [8]Gynecologic Oncology Center, Department of Obstetrics and Gynecology, First Faculty of Medicine, Charles University and General University Hospital, Prague

5278

Introduction

Epithelial ovarian carcinoma (EOC) is among the top five causes of cancer-related death in women. Most women with EOC achieve complete remission after primary or interval cytoreductive surgery combined with chemotherapy based on a platinum-taxane doublet. Homologous recombination (HR) defects imposed by germline or somatic BRCA1 DNA repair-associated (*BRCA1*) or *BRCA2* mutations are not only key determinants of platinum sensitivity in EOC patients but also provide a strong rationale for maintenance therapy based on poly(ADP-ribose) polymerase (PARP) inhibitors, which is generally associated with improved progression-free survival (PFS). Besides direct cytotoxic and cytostatic properties, PARPi have been shown to mediate multipronged immunostimulatory effects, largely reflecting their ability to inhibit DNA repair in malignant cells and indicating the possibility to synergy with immune-check point inhibitors (ICIs). Thus, combinatorial regimens involving PARPi and ICIs are emerging as a potential strategy for the management of EOC. Despite recent advances in the clinical evaluation of PARPi and ICIs therapy, the impact of combined therapy on anti-tumor immunity development, spatial contexture and ICI-responsive T cells subsets within tumor-microenvironment is relatively unknown in EOC.

Figure 1

PARPi drive a STING-dependent type I IFN signaling in *Brca1*$^{-/-}$ BR5 as compared to ID8 cell line.

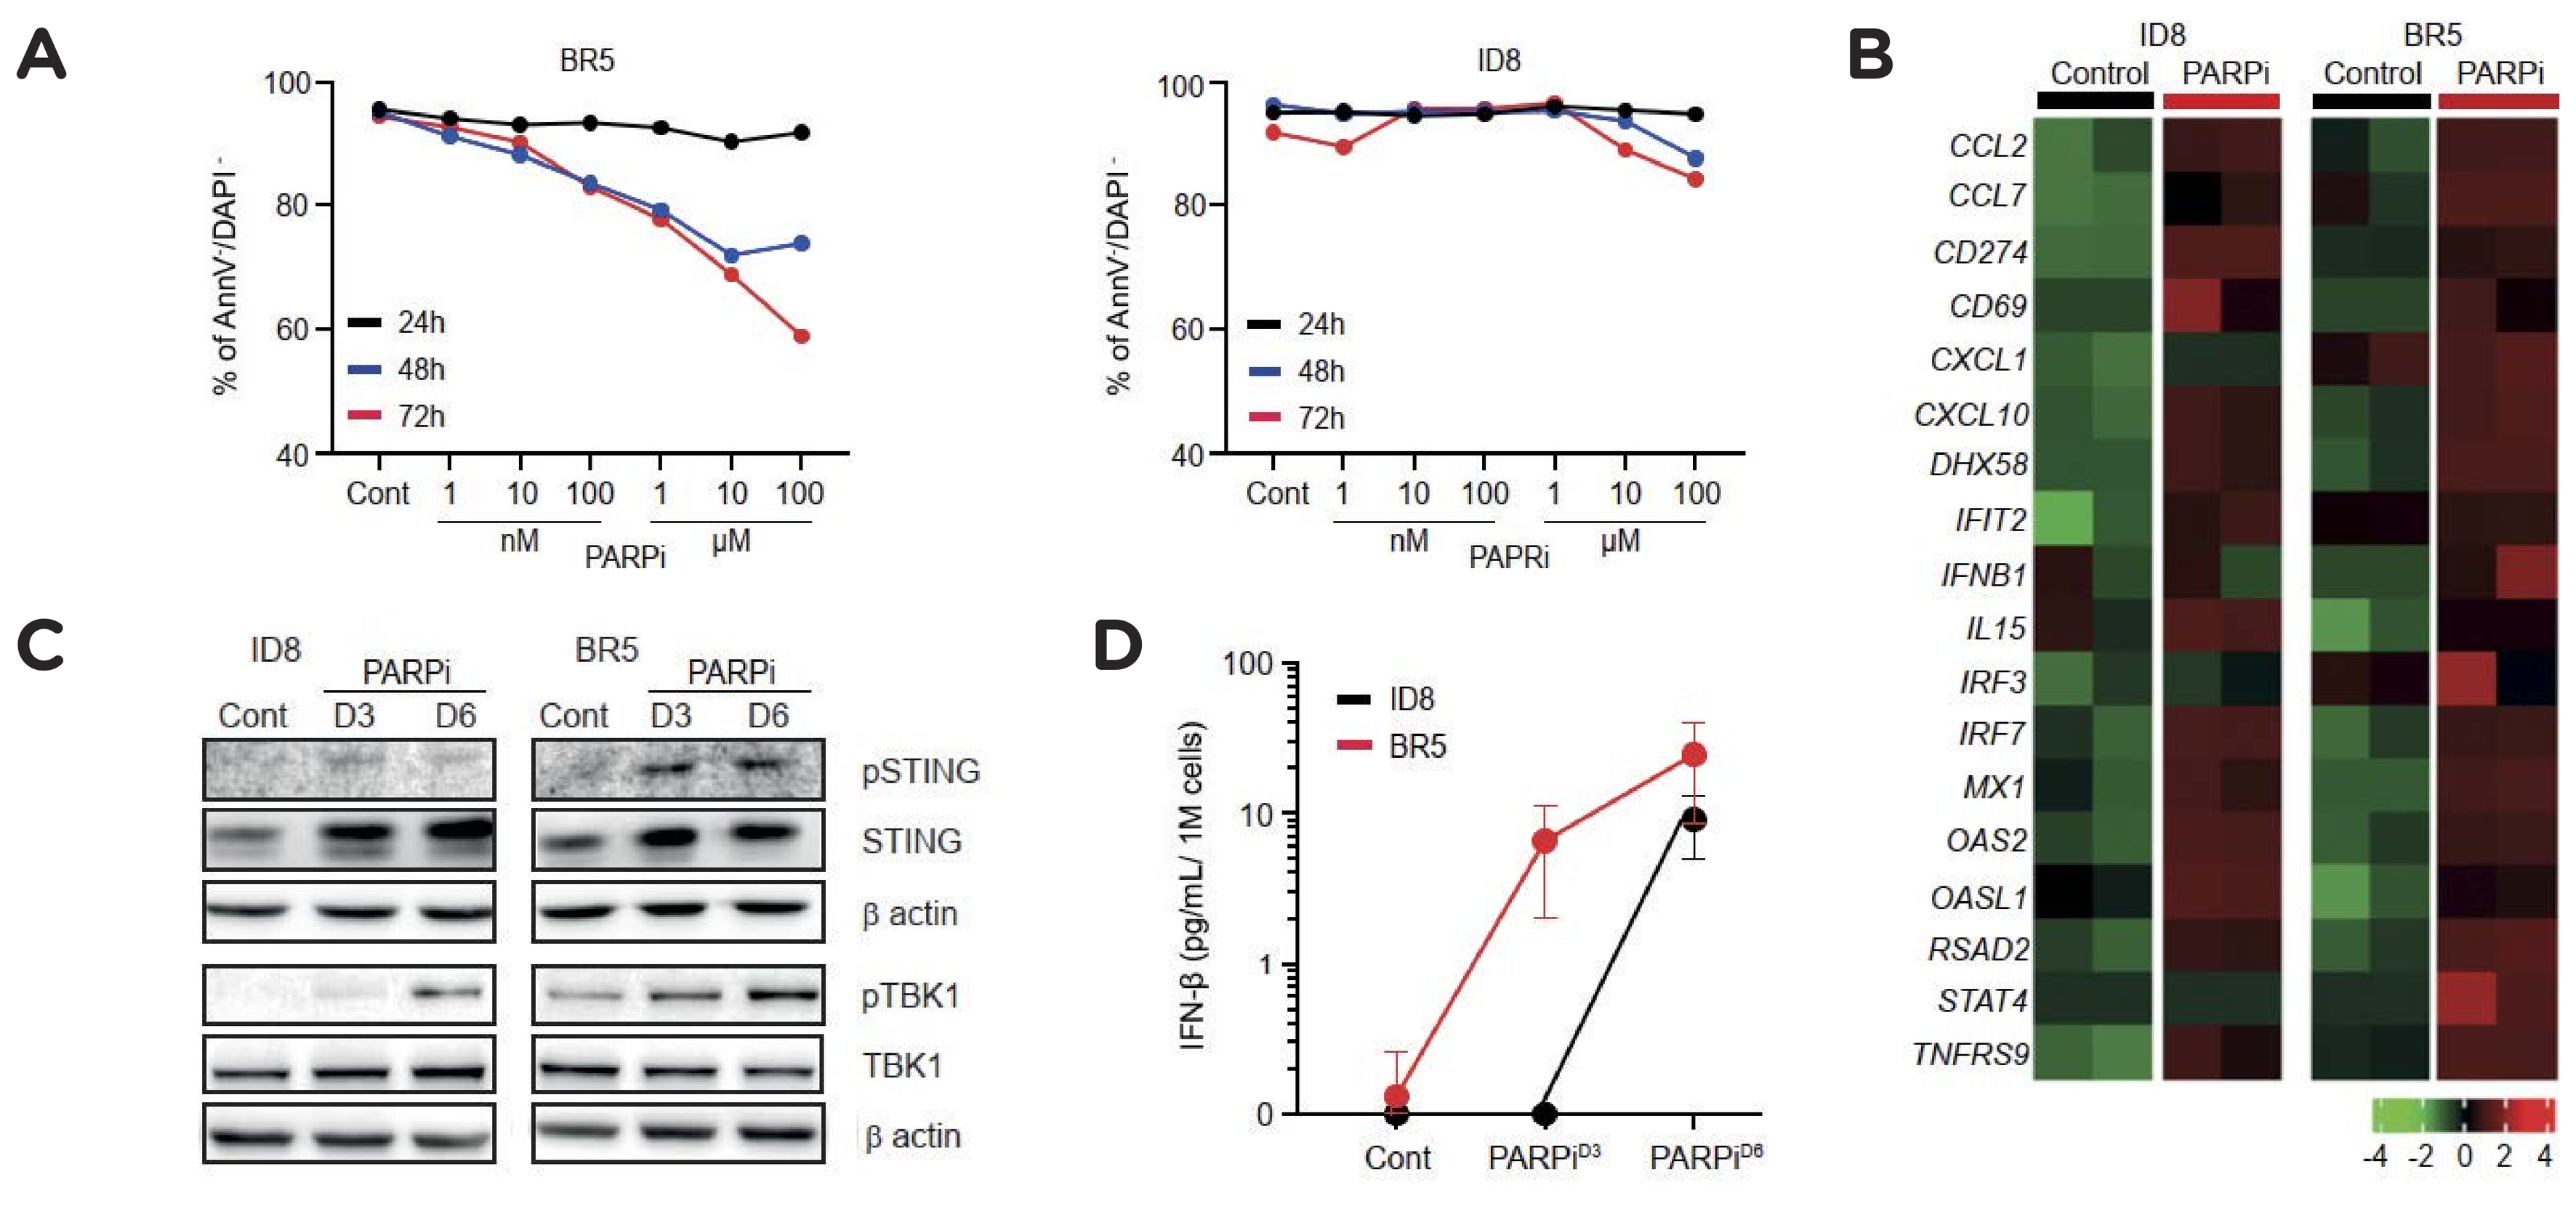

A
BR5
ID8
% of AnnV⁻/DAPI⁻
24h
48h
72h
Cont 1 10 100 1 10 100
nM PARPi μM
B
ID8 BR5
Control PARPi Control PARPi
CCL2
CCL7
CD274
CD69
CXCL1
CXCL10
DHX58
IFIT2
IFNB1
IL15
IRF3
IRF7
MX1
OAS2
OASL1
RSAD2
STAT4
TNFRS9
-4 -2 0 2 4
C
ID8 BR5
Cont D3 D6
pSTING
STING
β actin
pTBK1
TBK1
β actin
D
IFN-β (pg/mL/ 1M cells)
ID8
BR5
Cont PARPi^D3 PARPi^D6

(A) Flow cytometry analyses for frequency of viable AnnV⁻ and DAPI⁻ BR5 and ID8 cells after Olaparib (PARPi) at 24, 48 and 72 hours. **(B)** Supervised hierarchical clustering of gene signatures related to type I IFN signaling in control and 1μM Olaparib (PARPi)-treated ID8 and BR5 cell lines as determined by RNA sequencing (RNAseq). **(C)** Phosphorylation of STING and TBK1 in control and Olaparib (PARPi)-treated ID8 and BR5 cell lines at day 3 (D3) and 6 (D6), determined by immunoblotting. **(D)** IFNβ production by ID8 and BR5 cells 24 h after Olaparib (PARPi) therapy, determined by ELISA.

Aim of study

Here we evaluate PARPi immunomodulatory activity, with a specific focus on molecular and cellular pathways that can be harnessed to develop superior combinatorial regimens with ICIs for clinical management of EOC, using multiparametric flow cytometry, multiplex immunostaining, single cells transcriptomics and functional studies in experimental *Brca1*$^{-/-}$ BR5 ovarian syngeneic mouse model.

Figure 2

Figure 2. PARPi elicit STING-dependent anti-tumor immunity in *Brca1*$^{-/-}$ BR5 cell line.

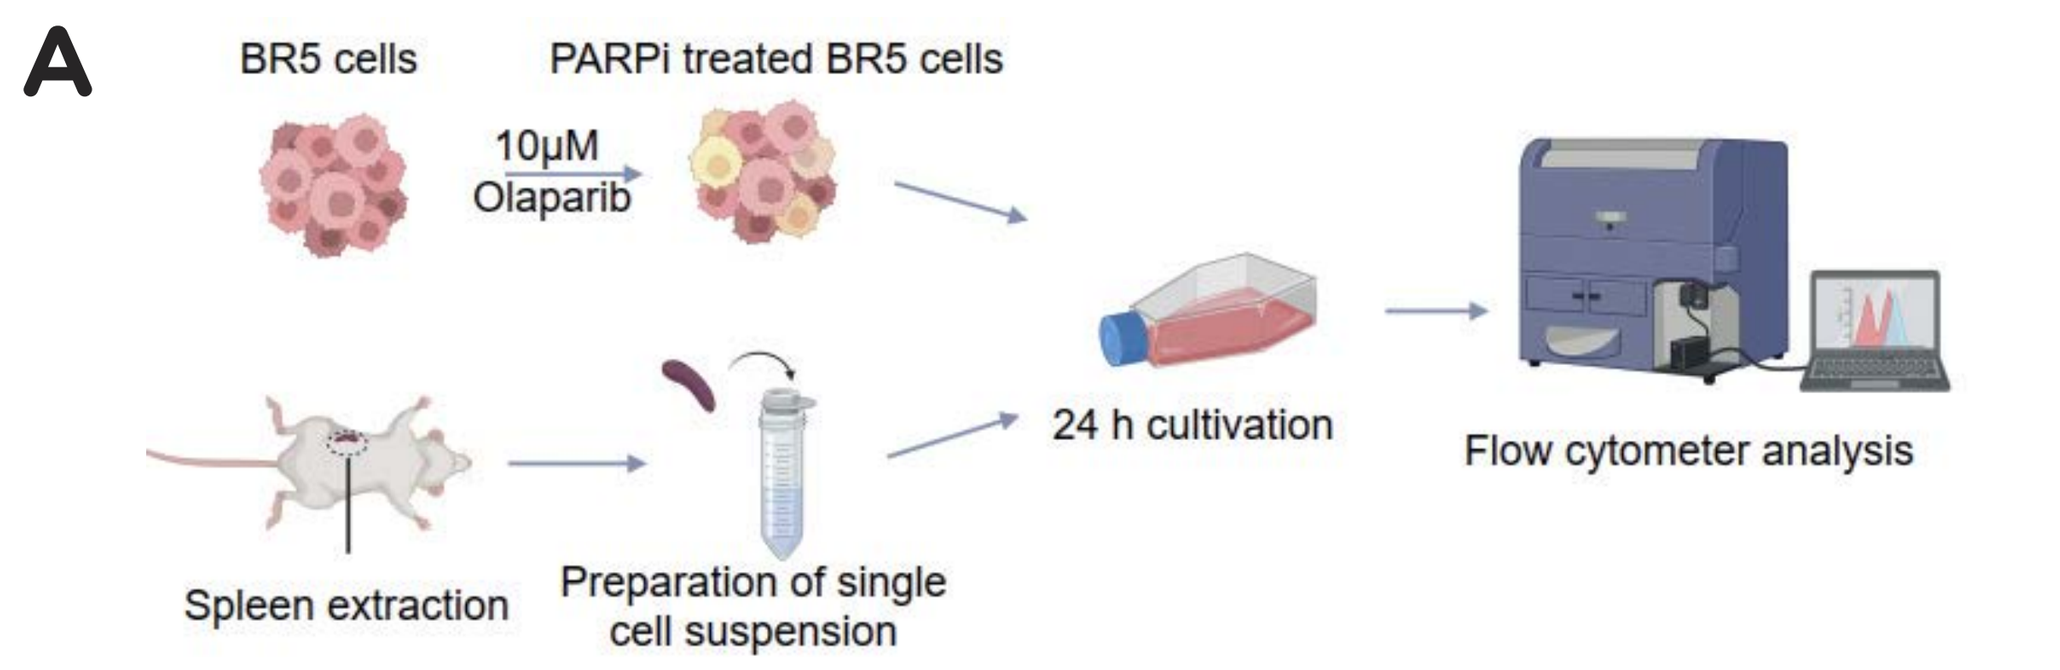

A
BR5 cells
PARPi treated BR5 cells
10μM Olaparib
24 h cultivation
Flow cytometer analysis
Spleen extraction
Preparation of single cell suspension

(A) Experimental design for the analysis of anti-tumor immunity development in BR5 mouse model. **(B, C)** Box plot showing the percentage of CD69⁺ and CD107⁺CD8⁺ T cells **(B)** and CD3⁻CD56⁺ NK cells **(C)** in mouse tumor samples. Box plots: lower quartile, median, upper quartile; whiskers, minimum, maximum. Statistical significance was calculated by the Mann-Whitney test. *p* values are indicated.

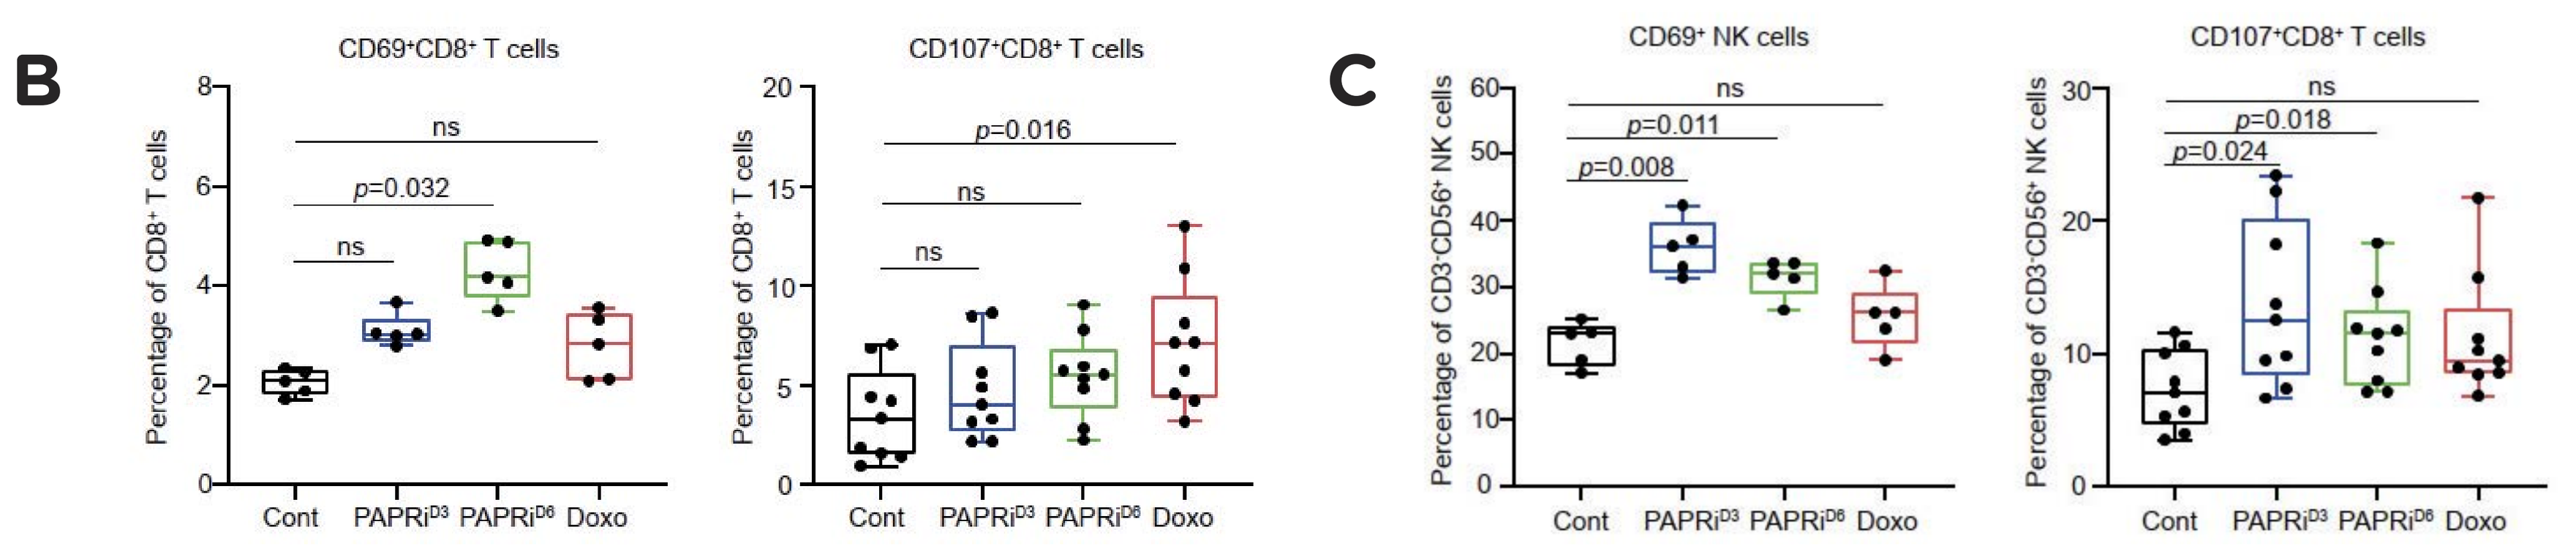

B
CD69⁺CD8⁺ T cells
CD107⁺CD8⁺ T cells
Percentage of CD8⁺ T cells
p=0.032
p=0.016
ns
Cont PAPRi^D3 PAPRi^D6 Doxo
C
CD69⁺ NK cells
CD107⁺CD8⁺ T cells
Percentage of CD3⁻CD56⁺ NK cells
p=0.011
p=0.008
p=0.018
p=0.024
ns

Figure 3

PARPi in both early and late setting provide survival benefit to *Brca1*$^{-/-}$ BR5 experimental model.

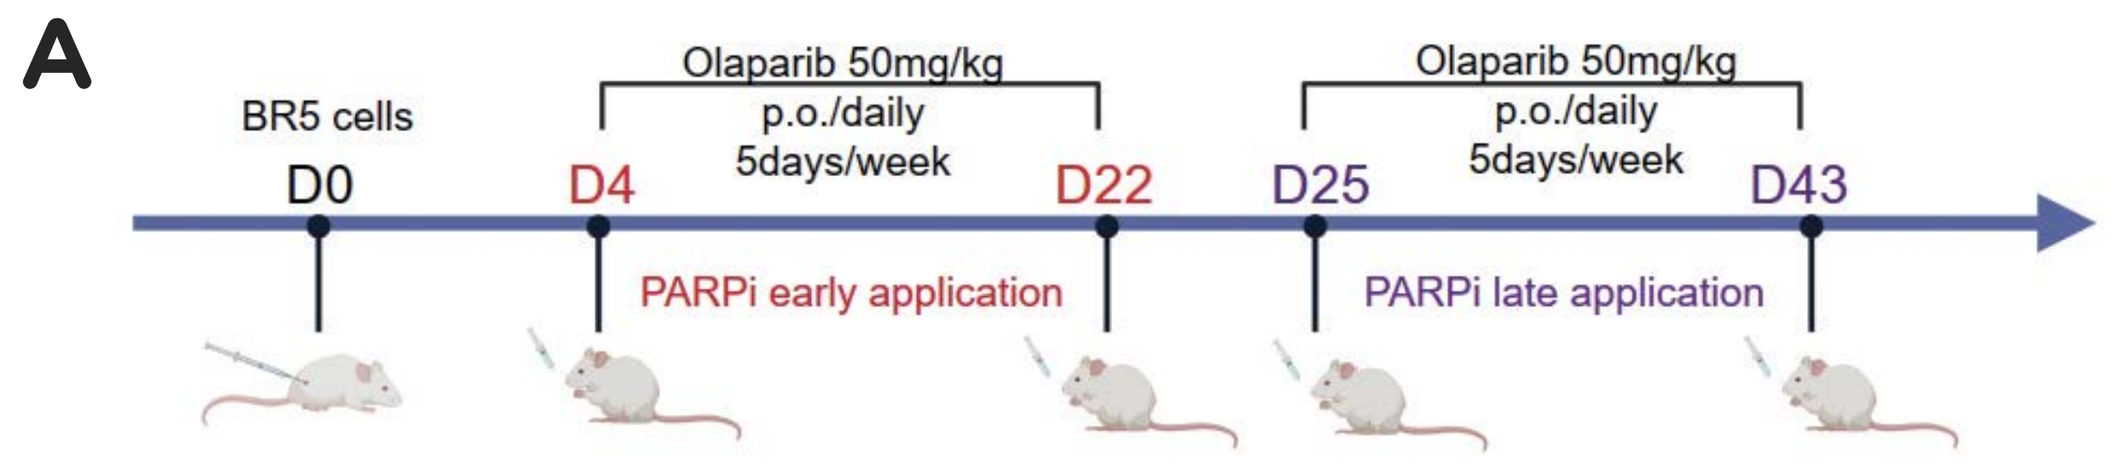

A
BR5 cells
D0
D4
D22
D25
D43
Olaparib 50mg/kg p.o./daily 5days/week
PARPi early application
PARPi late application

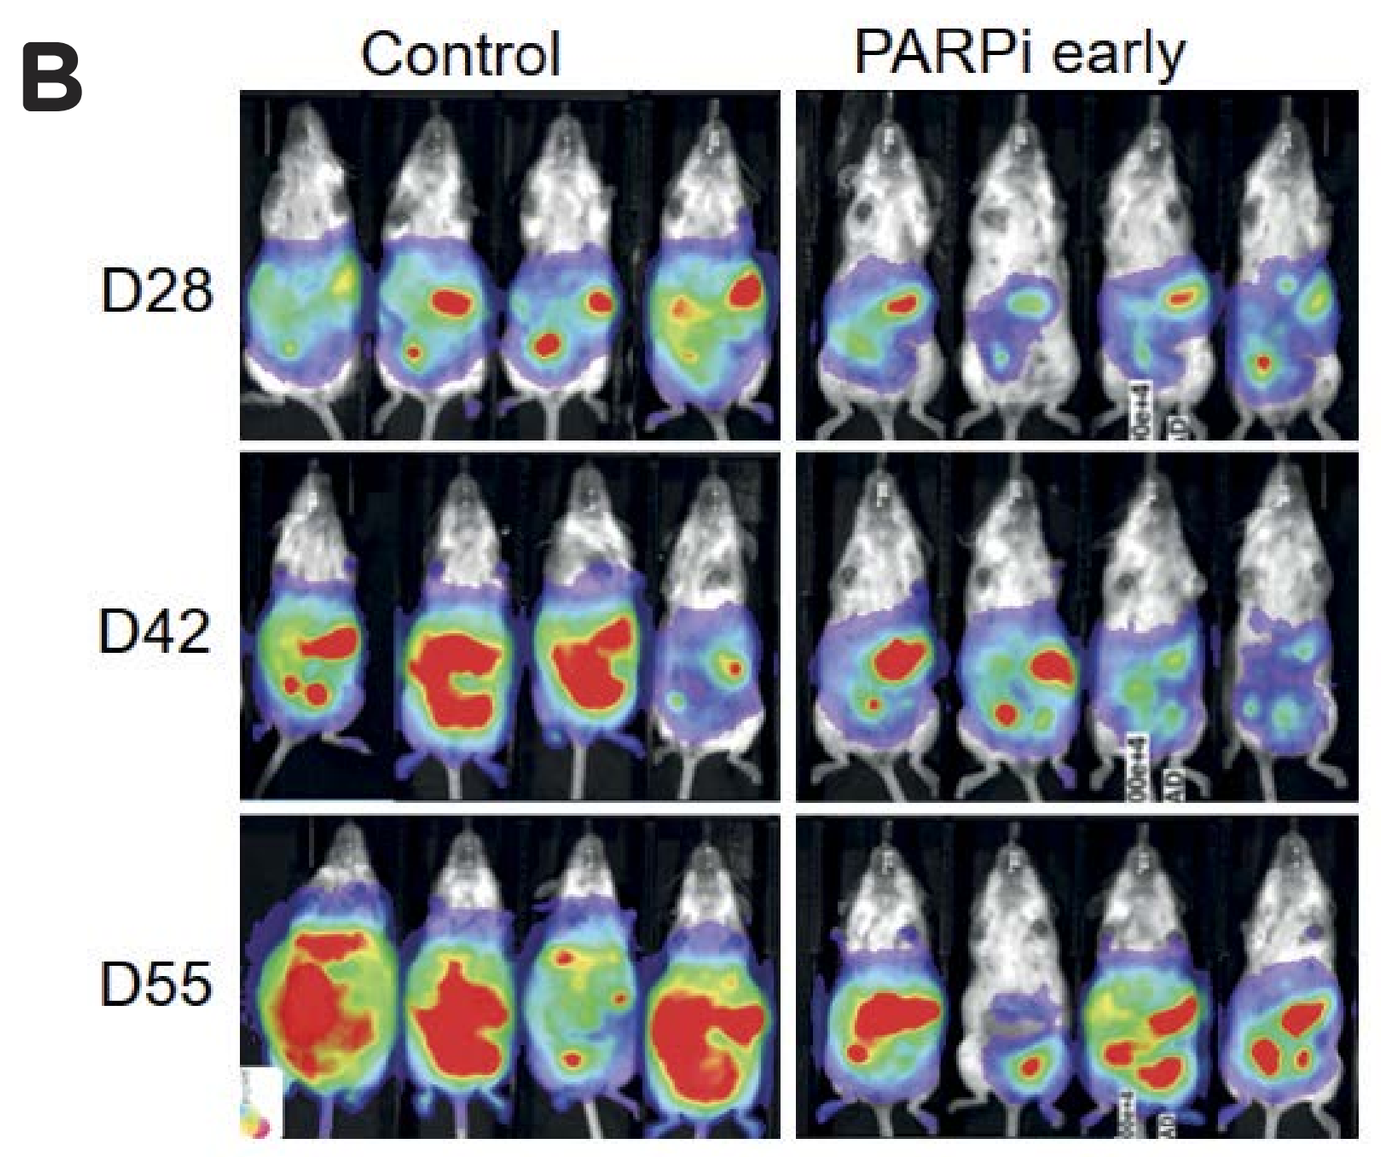

B
Control
PARPi early
D28
D42
D55

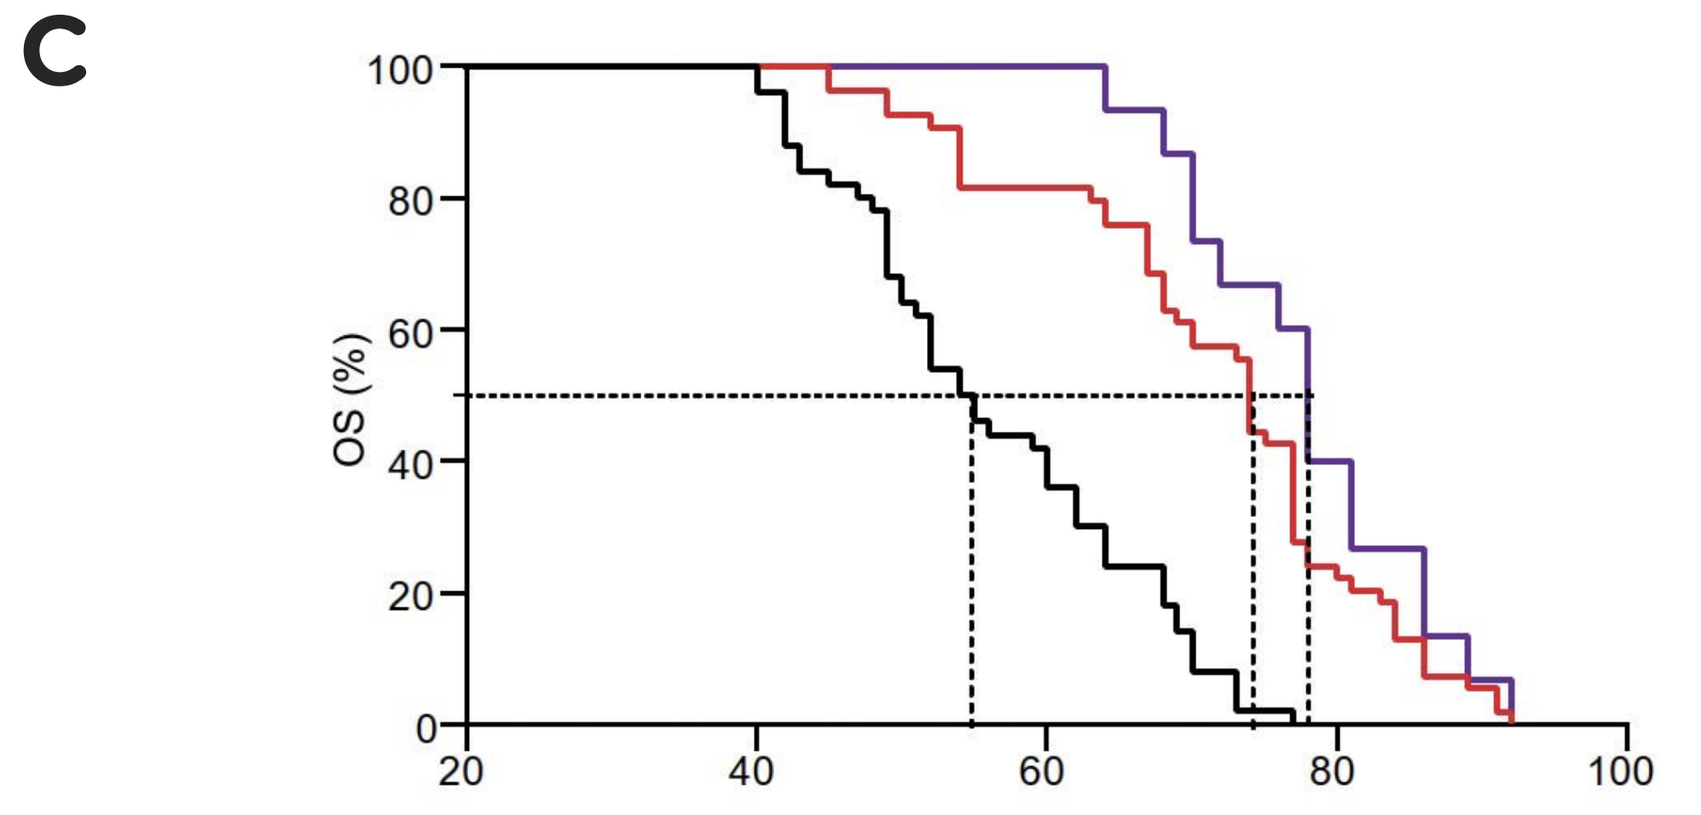

C
OS (%)

	Median OS	Control	PARPi early	PARPi late
Control	54.5	-	<0.0001	<0.0001
PARPi early	74	<0.0001	-	ns
PARPi late	78	<0.0001	ns	-

(A) Experimental design and **(B)** representative bioluminescence images of early and late Olaparib (PARPi) treatment in BR5 model. **(C)** Overall survival (OS) of BR5 experimental model after Olaparib (PARPi) early and late application. Survival curve was estimated by the Kaplan-Meier method and differences between groups were evaluated using log-rank test.

Figure 4

PARPi enhance the clinical benefit of ICIs therapy in *Brca1*$^{-/-}$ BR5 experimental model.

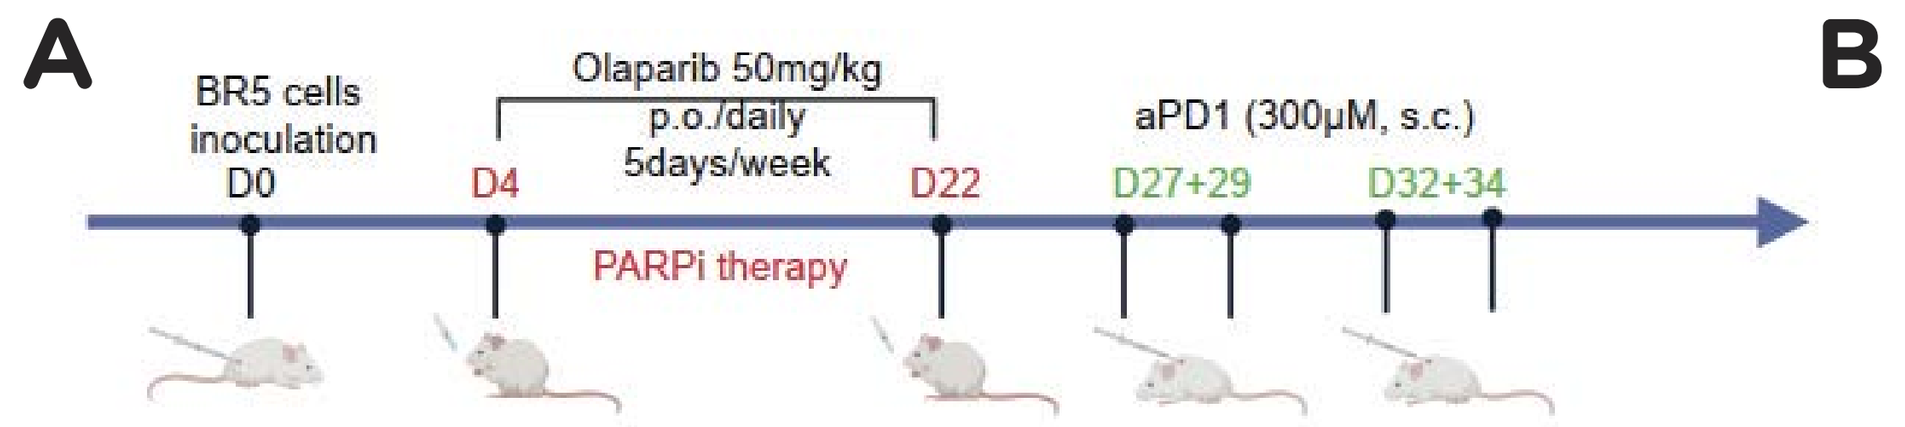

A
BR5 cells inoculation
D0
D4
D22
D27+29
D32+34
Olaparib 50mg/kg p.o./daily 5days/week
aPD1 (300μM, s.c.)
PARPi therapy

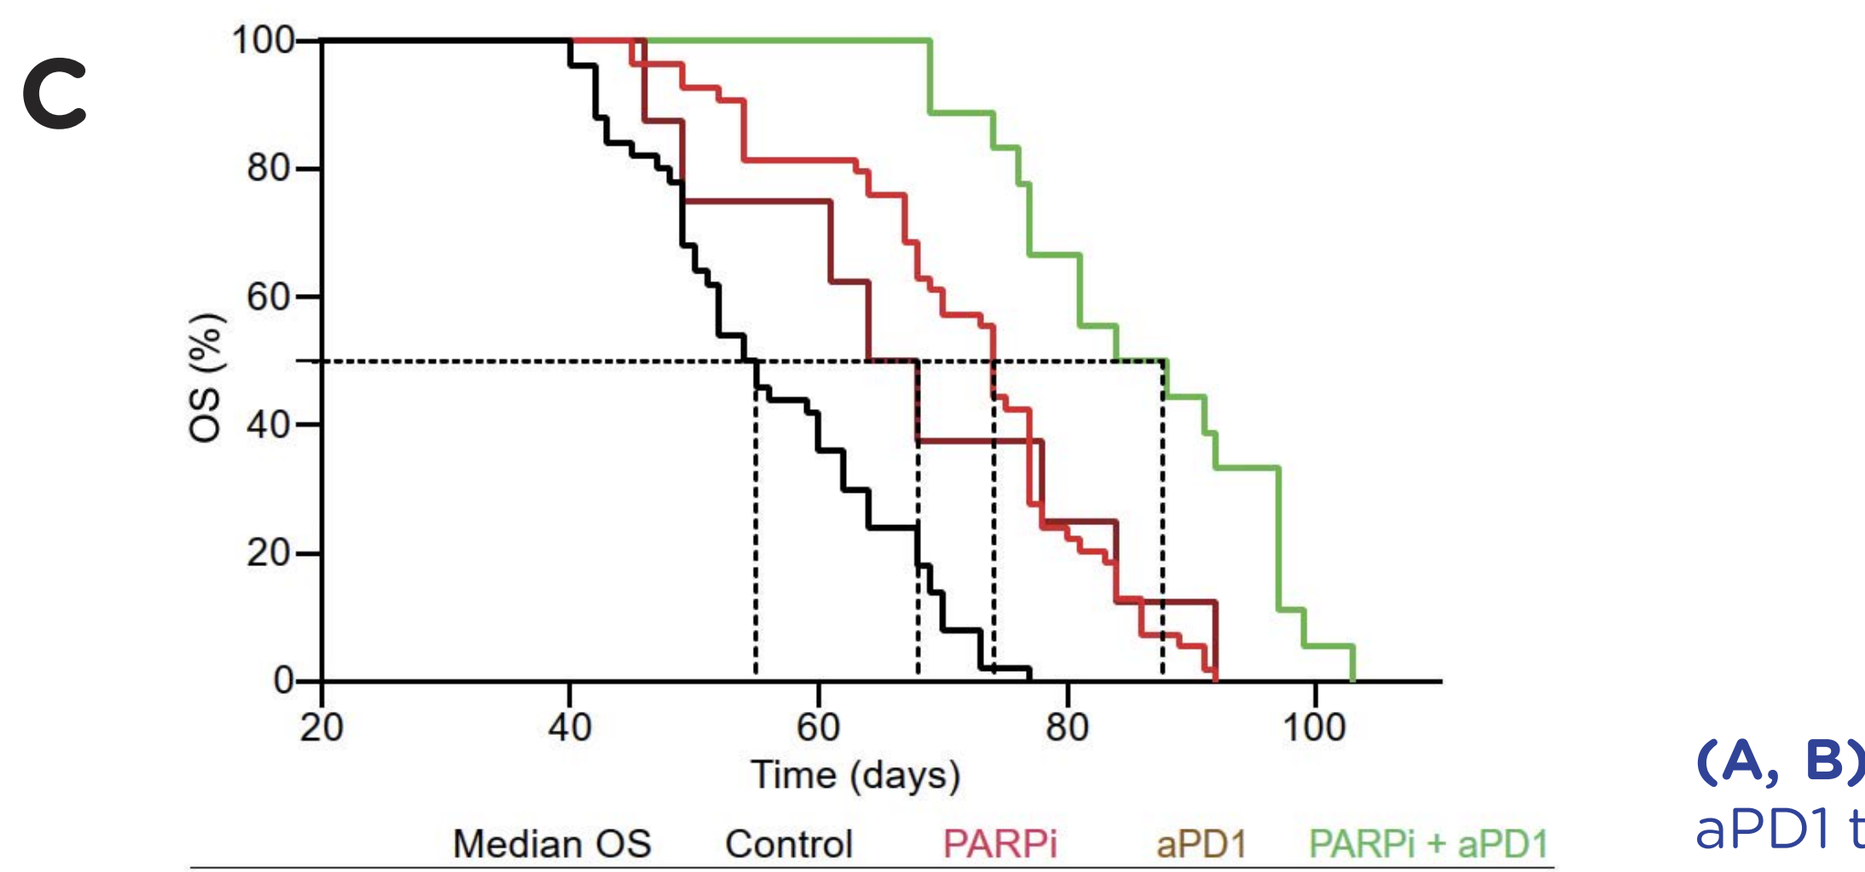

C
OS (%)
Time (days)

	Median OS	Control	PARPi	aPD1	PARPi + aPD1
Control	54.5	-	<0.0001	0.0128	<0.0001
PARPi	74	<0.0001	-	ns	<0.0001
aPD1	66	0.0128	ns	-	0.0061
PARPi + aPD1	86	<0.0001	<0.0001	0.0061	-

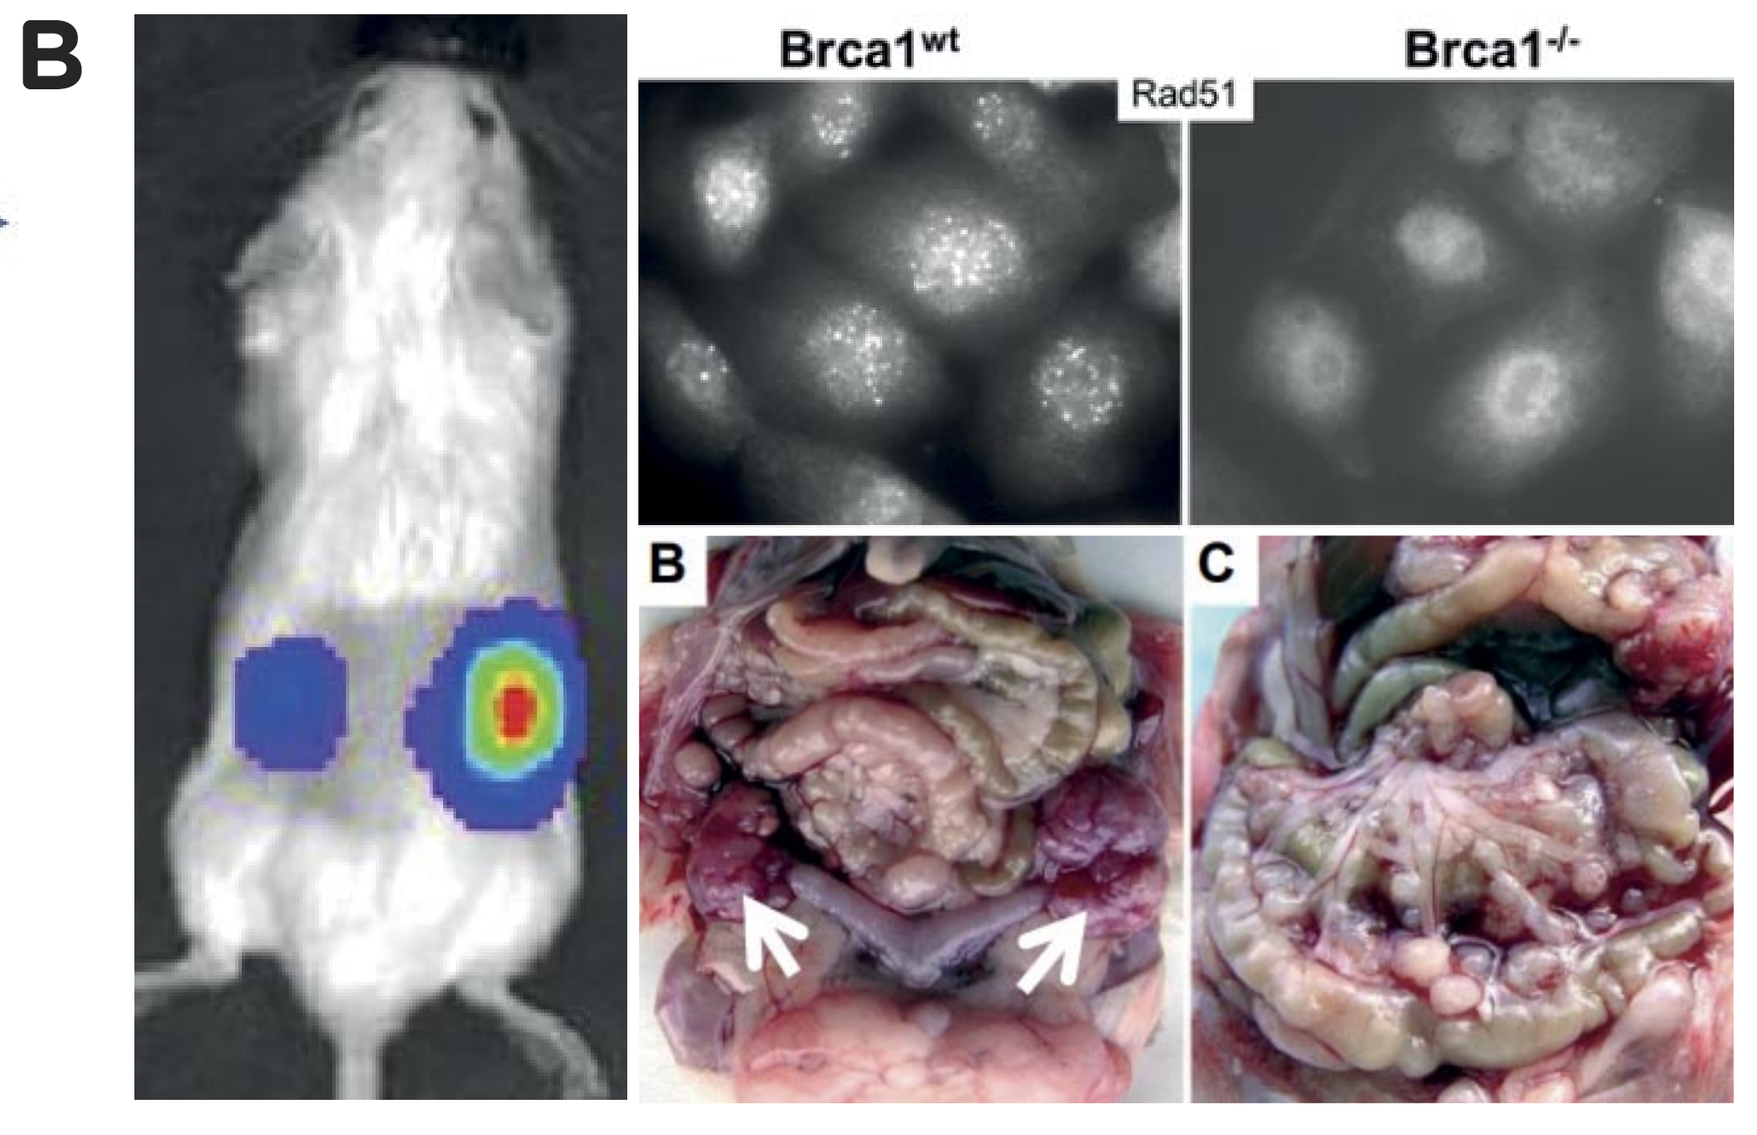

B
Brca1^wt
Brca1^-/-
Rad51
B
C

(A, B) Experimental design of combined Olaparib (PARPi) and aPD1 therapy in syngeneic BR5 mouse model. **(C)** Overal Survival (OS) of BR5 experimental model after combined Olaparib (PARPi) and aPD1 therapy as compared to monotherapy. Survival curve was estimated by the Kaplan-Meier method and differences between groups were evaluated using log-rank test.

Figure 5

Immunomodulatory properties of combined PARPi and aPD1 therapy. The combination of PARPi and aPD1 positively regulate the balance between adaptive anti-tumor immunity and innate myeloid components and improve the cytotoxic T cell profile in *Brca1*$^{-/-}$ BR5 experimental mouse model.

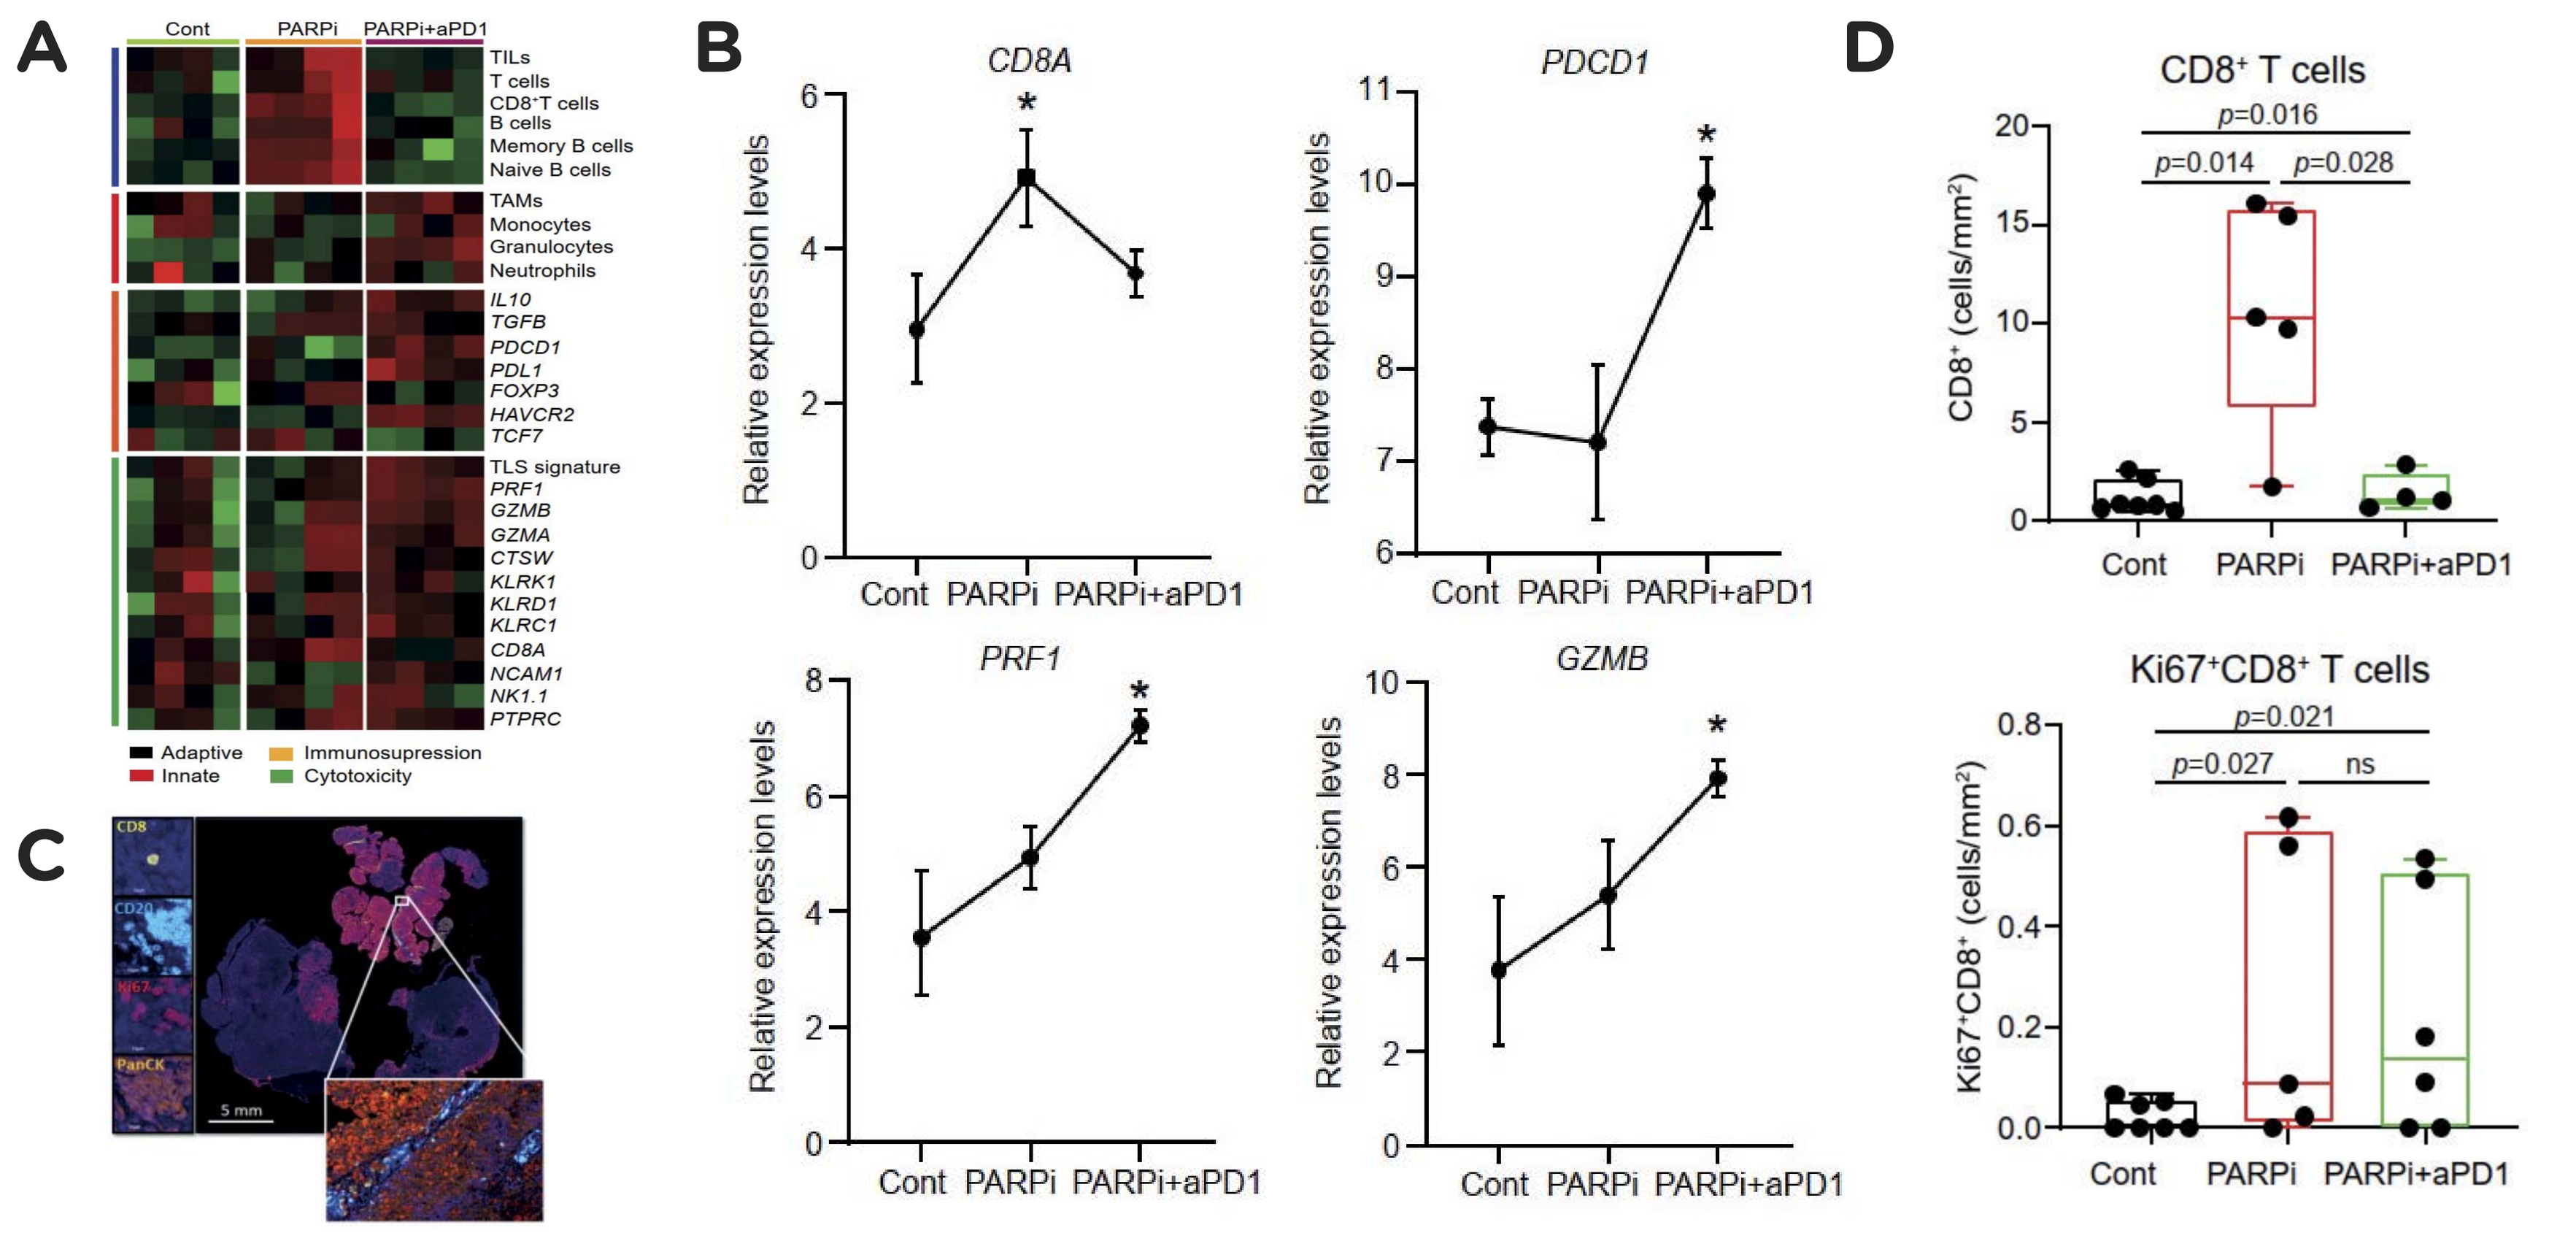

A
Cont PARPi PARPi+aPD1
TILs
T cells
CD8⁺T cells
B cells
Memory B cells
Naive B cells
TAMs
Monocytes
Granulocytes
Neutrophils
IL10
TGFB
PDCD1
PDL1
FOXP3
HAVCR2
TCF7
TLS signature
PRF1
GZMB
GZMA
CTSW
KLRK1
KLRD1
KLRC1
CD8A
NCAM1
NK1.1
PTPRC
Adaptive
Innate
Immunosupression
Cytotoxicity
B
CD8A
PDCD1
PRF1
GZMB
Relative expression levels
Cont PARPi PARPi+aPD1
C
CD8
CD20
Ki67
PanCK
5 mm
D
CD8⁺ T cells
CD8⁺ (cells/mm²)
p=0.016
p=0.014
p=0.028
Ki67⁺CD8⁺ T cells
Ki67⁺CD8⁺ (cells/mm²)
p=0.021
p=0.027
ns
Cont PARPi PARPi+aPD1

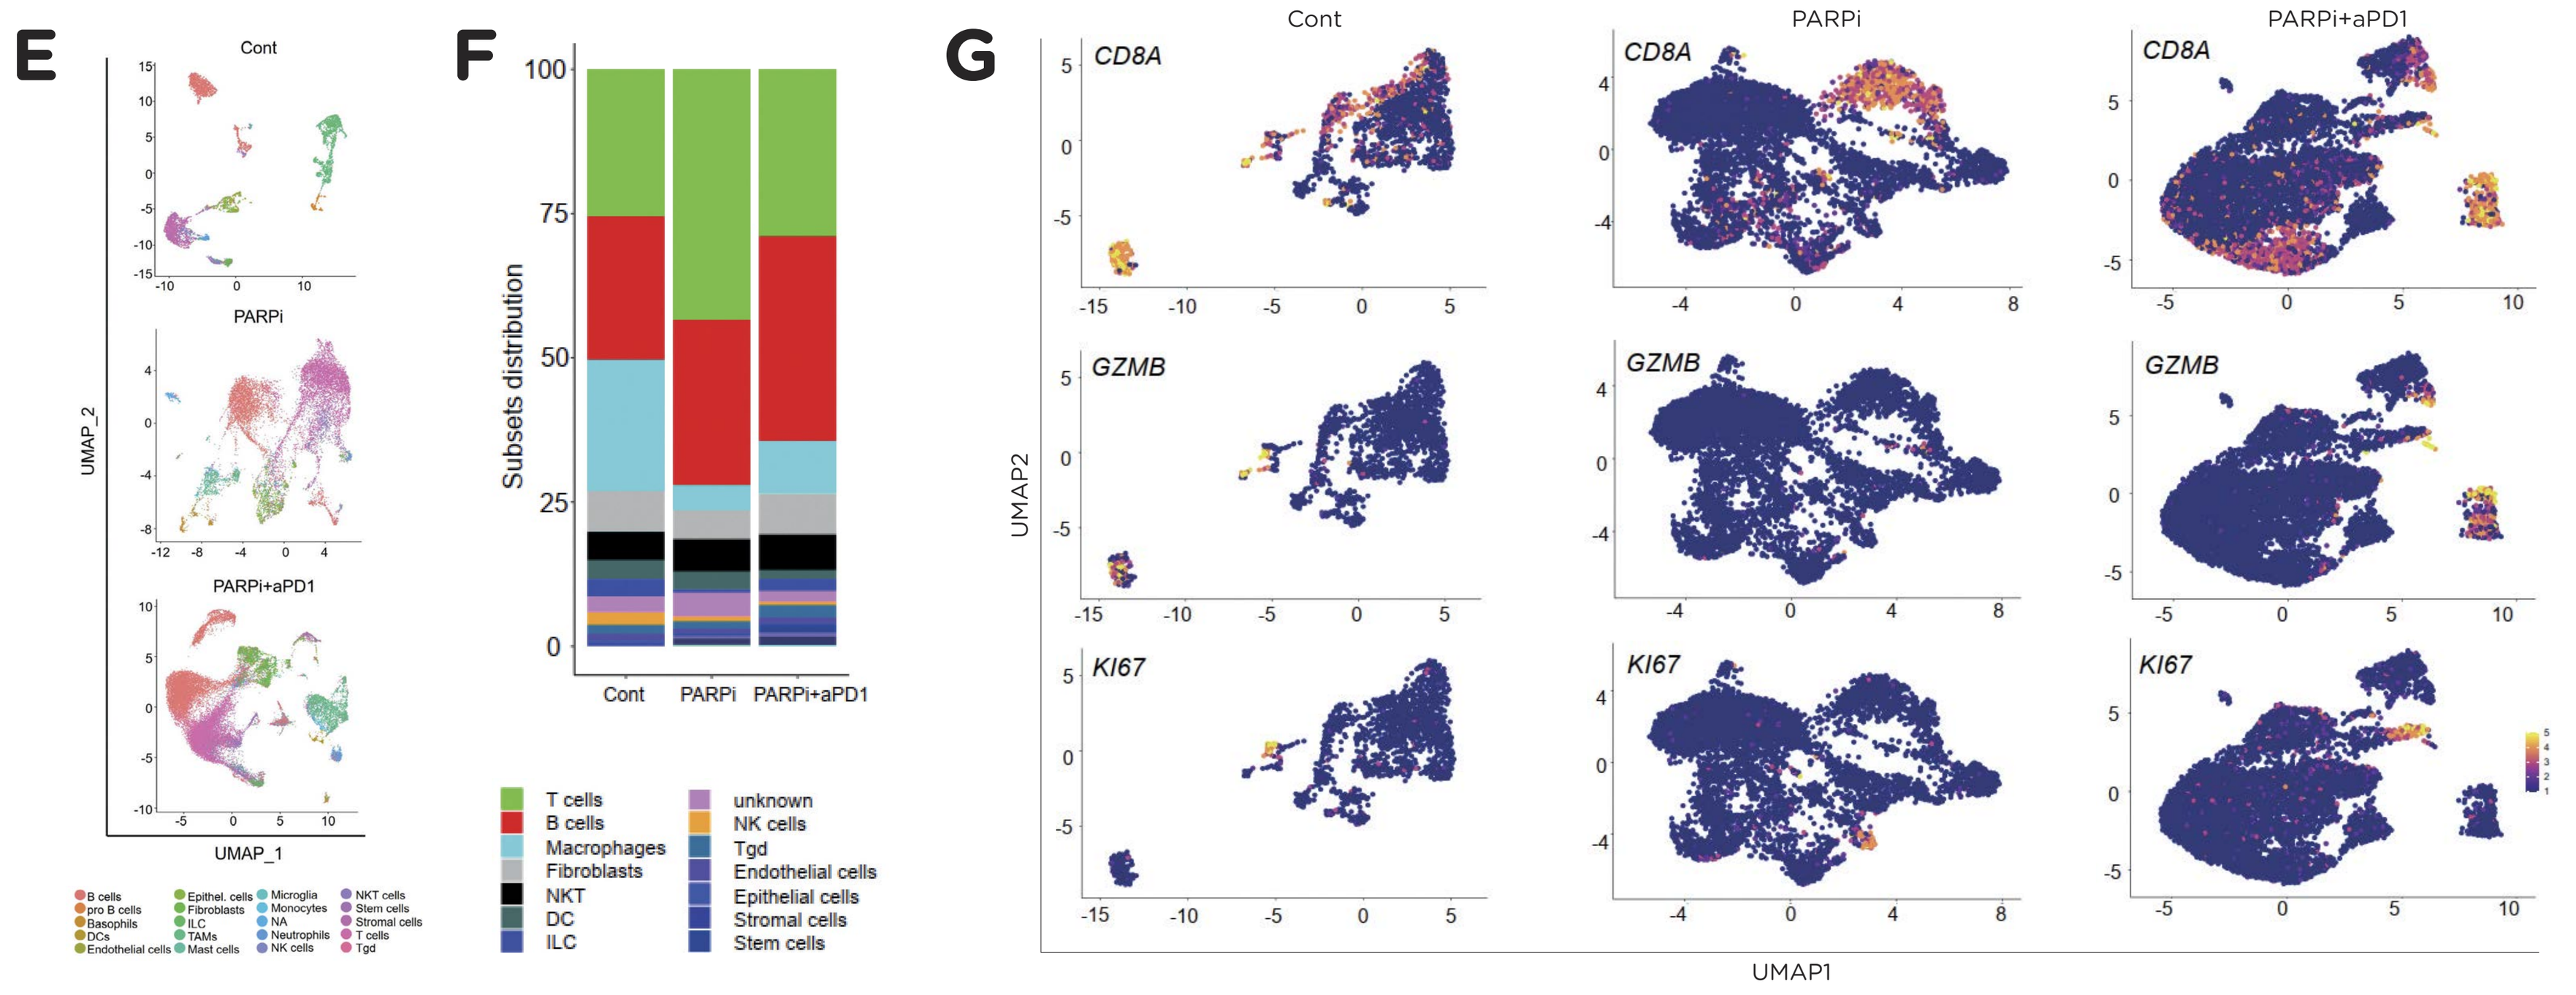

E
Cont
PARPi
PARPi+aPD1
UMAP_1
UMAP_2
F
Subsets distribution
Cont PARPi PARPi+aPD1
T cells
B cells
Macrophages
Fibroblasts
NKT
DC
ILC
unknown
NK cells
Tgd
Endothelial cells
Epithelial cells
Stromal cells
Stem cells
G
Cont
PARPi
PARPi+aPD1
CD8A
GZMB
KI67
UMAP1
UMAP2

(A) Supervised hierarchical clustering of gene signatures related to adaptive (blue) and innate (red) immunity, immunosuppression (orange) and cytotoxicity (green) and **(B)** gene expression signature associated with CD8⁺ T cells (determined by MCP counter) and relative gene expression levels of *CD8A, GZMB, PRF1, PDCD1* as determined by RNAseq data in tumor samples of *Brca1*$^{-/-}$ BR5 model after combined PARPi and aPD1 as compared to PARPi alone. **(C)** Representative image of CD8, CD20, Ki67 and PanCK determined by immunostaining. Scale bars 5mm and 10 μm. **(D)** Density of CD8⁺ T cells and Ki67⁺ CD8⁺ T cells in tumor samples of *Brca1*$^{-/-}$ BR5 model after combined PARPi and aPD1 as compared to PARPi alone. **(E)** t-distributed stochastic neighbour embedding (tSNE) analysis of cell that passed quality checks in *Brca1*$^{-/-}$ BR5 tumor after combined PARPi and aPD1 as compared to PARPi alone. Cells and clusters are color coded by the major cell type found. **(F, G)** The distribution of cell types **(F)** and feature plot showing expression of key genes across CD8⁺ T cell clusters **(G)** between control, PARPi and combined PARPi and aPD1 therapy. Statistical significance was calculated by the Mann-Whitney test. *p* values are indicated.

Conclusions

Our finding demonstrates, that PARPi elicit STING-dependent anti-tumor immunity in *Brca1*$^{-/-}$ BR5 derived experimental model as compared to *BRCA1* proficient ID8 model. Supporting this notion, the combination of PARPi and aPD1 positively regulate the balance between adaptive anti-tumor immunity and innate myeloid components and significantly improve the cytotoxic T cell profile as shown in both experimental mice model. We surmise that rationally designed combinations of PARPi and immunotherapy might be critical to unlock immunosuppression in the EOC microenvironment in support of clinical efficacy.

For more information please contact: holicek@sotio.com, fucikova@sotio.com